HCP EDUCATION 660

Addressing Barriers to Adopting First-line Standards of Care in la/mUC: A Community Practice-informed Initiative

Authors: Kia Jones, PhD[1]; Tariqa Ackbarali, PhD, MS, CHCP[1]; Petros Grivas, MD, PhD, FASCO[2]; Neal D. Shore, MD, FACS[3]

Affiliation: [1]MedliveCME, Needham, MA; [2]University of Washington and Fred Hutch Cancer Center, Seattle, WA; [3]START Carolinas/Carolina Urologic Research Center, Myrtle Beach, SC

This activity was supported by an educational grant from Astellas and Pfizer Inc.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® or the author of this poster.

INTRODUCTION

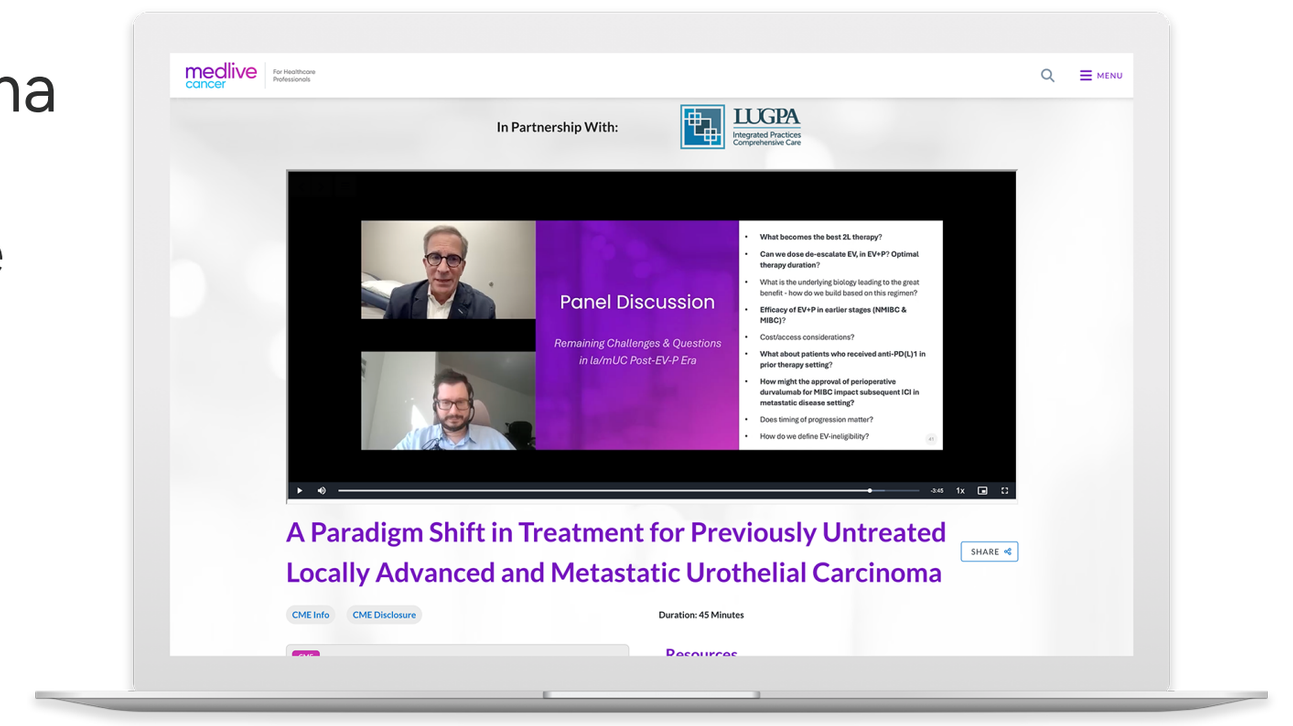
Panel Discussion
A Paradigm Shift in Treatment for Previously Untreated Locally Advanced and Metastatic Urothelial Carcinoma

Rapid advances in first-line (1L) therapy for locally advanced/metastatic urothelial carcinoma (la/mUC) have shifted standards and offered new opportunities to improve outcomes. These advances also introduce complexities in clinical decision-making, including eligibility, sequencing, and adverse-event (AE) management. In the community setting, these challenges are amplified by variations in resources, infrastructure, and care coordination. To address these challenges and support adoption of evolving standards of care, a targeted educational initiative was developed for GU oncology teams.

METHODOLOGY

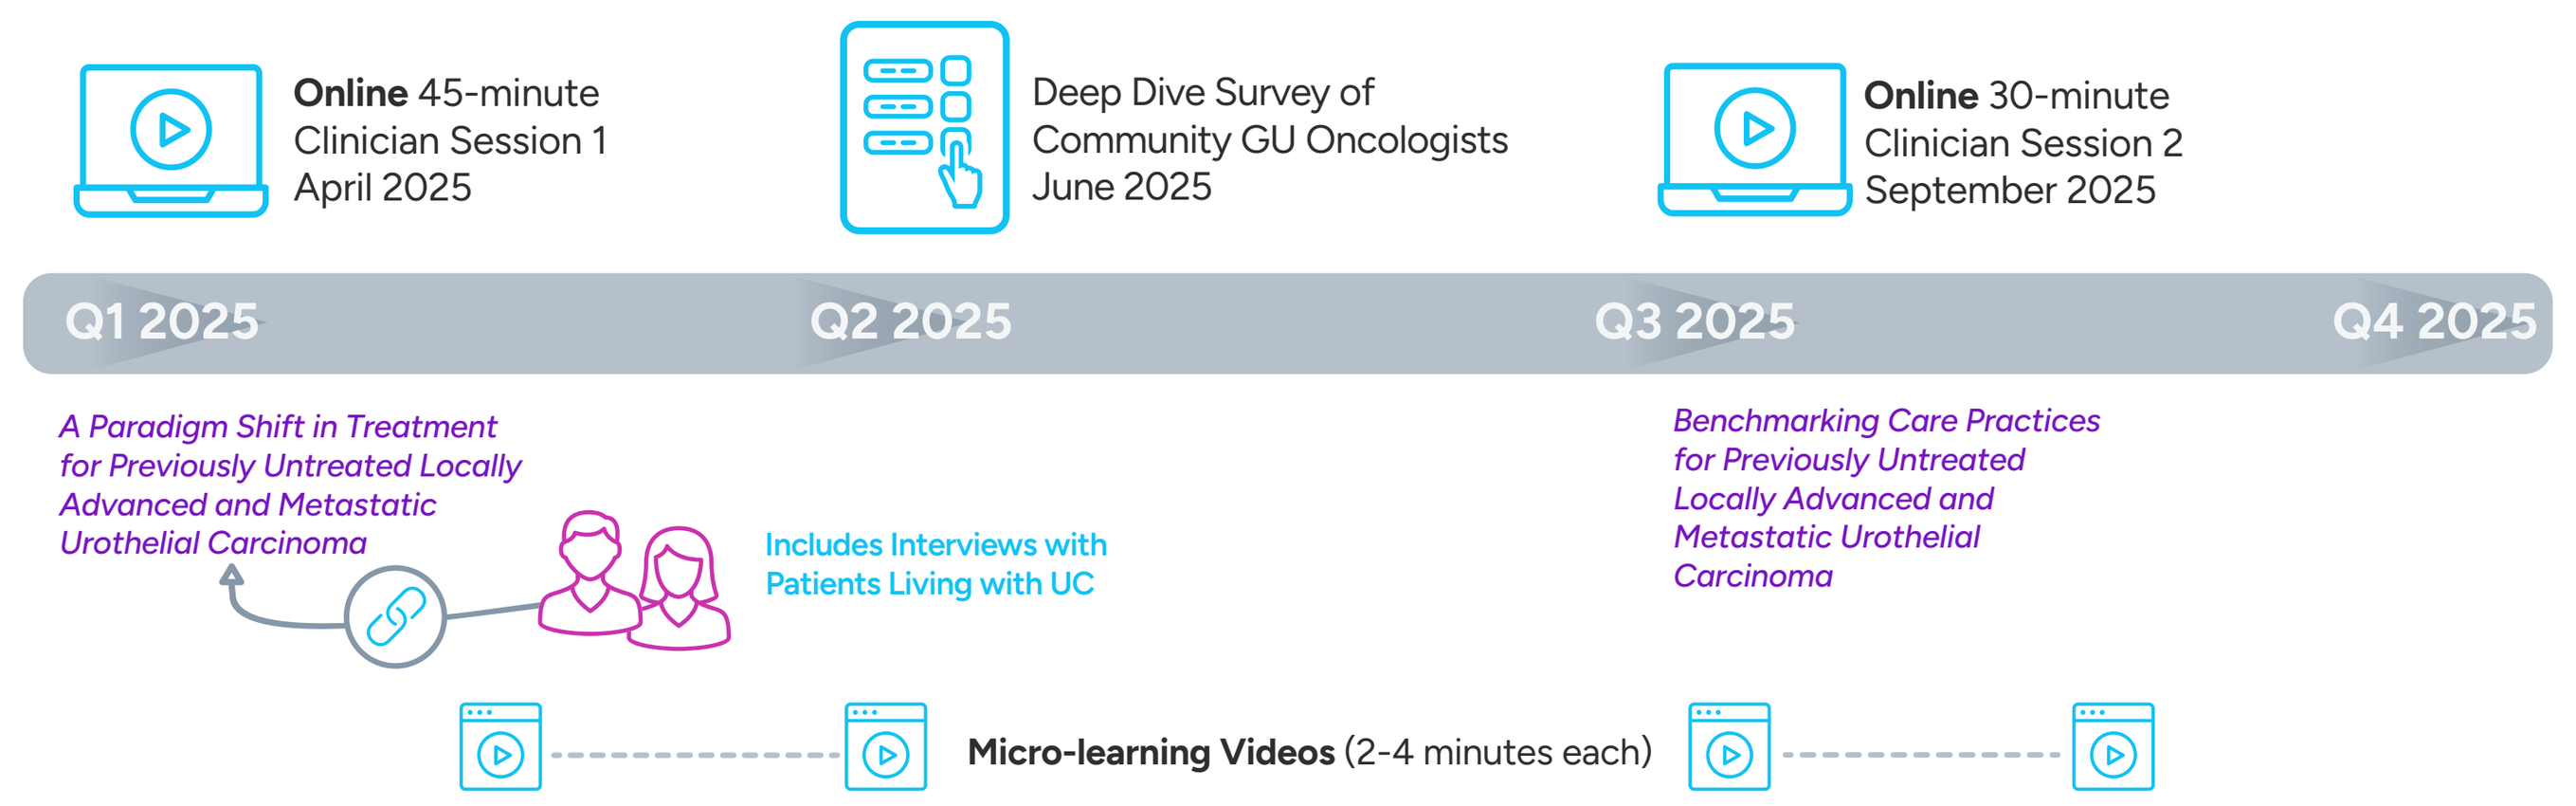
Online 45-minute Clinician Session 1 April 2025
Deep Dive Survey of Community GU Oncologists June 2025
Online 30-minute Clinician Session 2 September 2025
Q1 2025
Q2 2025
Q3 2025
Q4 2025
A Paradigm Shift in Treatment for Previously Untreated Locally Advanced and Metastatic Urothelial Carcinoma
Includes Interviews with Patients Living with UC
Benchmarking Care Practices for Previously Untreated Locally Advanced and Metastatic Urothelial Carcinoma
Micro-learning Videos (2-4 minutes each)

Enduring CME activities available through April 2026

Educational Program and Evaluation Details

Interventions

Two certified (1.25 credits total) enduring sessions featured expert-panel education, videos, PDF resources, and slides. Primarily intended for physicians, nurse practitioners, and physician assistants in medical oncology and urology, and secondarily intended for nurses in oncology and urology. CME was augmented by targeted micro-learning videos via LinkedIn to NPI-verified oncologists.

Session 1: A Paradigm Shift in Treatment for Previously Untreated Locally Advanced and Metastatic Urothelial Carcinoma; launched April 29, 2025

Session 2: Benchmarking Care Practices for Previously Untreated Locally and Advanced Metastatic Urothelial Carcinoma; launched September 24, 2025

Formative (PPA survey): Email survey distributed via Sermo to oncology and urology clinicians treating people with UC; identified as treaters based on ICD-10 codes.

Data Collected

Engagement (CME participants, learners, and completers; micro-learning completers), changes in knowledge and competence, present attitudes and practice behaviors, intended practice changes, self-reported practice changes

Measurements

A survey-based approach measured impact of the CME as aligned with stated learning objectives via pre- and post-activity knowledge/competence questions developed in accordance with National Board of Medical Examiner guidelines,[1] a post-activity evaluation (inclusive of intended practice changes: open-ended), and a 60-day follow-up survey. Statistical tests of significance were applied to comparisons of individual questions.

Reference:

1. NBME Item-Writing Guide. Constructing written test questions for the health sciences. Nov 2020. www.nbme.org/item-writing-guide.

SURVEY RESULTS

Medline First-line mUC Survey: Demographics

95 clinicians responded to the practice-pattern survey

Average number of la/mUC patients managed per month

≤10 patients	11-20 patients	21-40 patients	>40 patients
19%	45%	24%	12%

Most survey respondents were community based, primarily medical oncologists and urologists, and managed a moderate to high volume of la/mUC patients

% of respondents	Specialty	Community based	Academic based
40%	Medical Oncologists	79%	21%
38%	Urologists	75%	25%
13%	Oncology NPs	67%	33%
9%	Oncology PAs	89%	11%

Medlive Survey Insights: Alignment with Guidelines (Cis-eligible)

Data indicated cisplatin eligibility as the most influential factor guiding 1L treatment selection.

- 37% prefer gem/cis + nivolumab + maintenance nivolumab
- 34% prefer gem/cis + maintenance avelumab
- 22% prefer enfortumab vedotin + pembrolizumab

71% preferred platinum-based chemo ± IO

Implications:

- Continued preference for platinum-based chemo + ICI likely reflects practice inertia and clinician familiarity, reinforced by their widespread use in other tumor types

Medlive Survey Insights: AE Management Challenges

Barriers in adopting ADC + IO therapy in 1L la/mUC	Percentage
Monitoring and managing side effects	56%
Reimbursement/coverage	43%
Determining patient eligibility	36%
Drug access/supply	33%

56% Cited **monitoring and managing side effects** as the biggest barrier to adopting ADC + IO regimen in practice

EDUCATIONAL IMPACT

Clinician's Changes in Knowledge

Approximately six months after the release of the first CME session

1,826 Participants
287 CME and
1,539 micro-learners

93% of participants designated their specialty as oncology/urology

71% of CME learners completed the session.

70% identified as current treaters who see an average of 14 patients with UC each month

Intended practice changes for patients with la/mUC (n=127) (Select all that apply)

	Percentage
Incorporate enfortumab vedotin + pembrolizumab as a 1L option for eligible patients with la/mUC	42%
Enhance efforts to stay current with pivotal trial data and evolving standards of care in 1L UC	32%
Re-evaluate the role of platinum-based chemotherapy regimens in light of updated guidelines and clinical data favoring ADC+IO combinations	32%
Improve patient counseling on treatment-related toxicities and side-effect management	26%

Oncology and urology learners demonstrated substantive pre- vs post-activity learning and confidence gains.

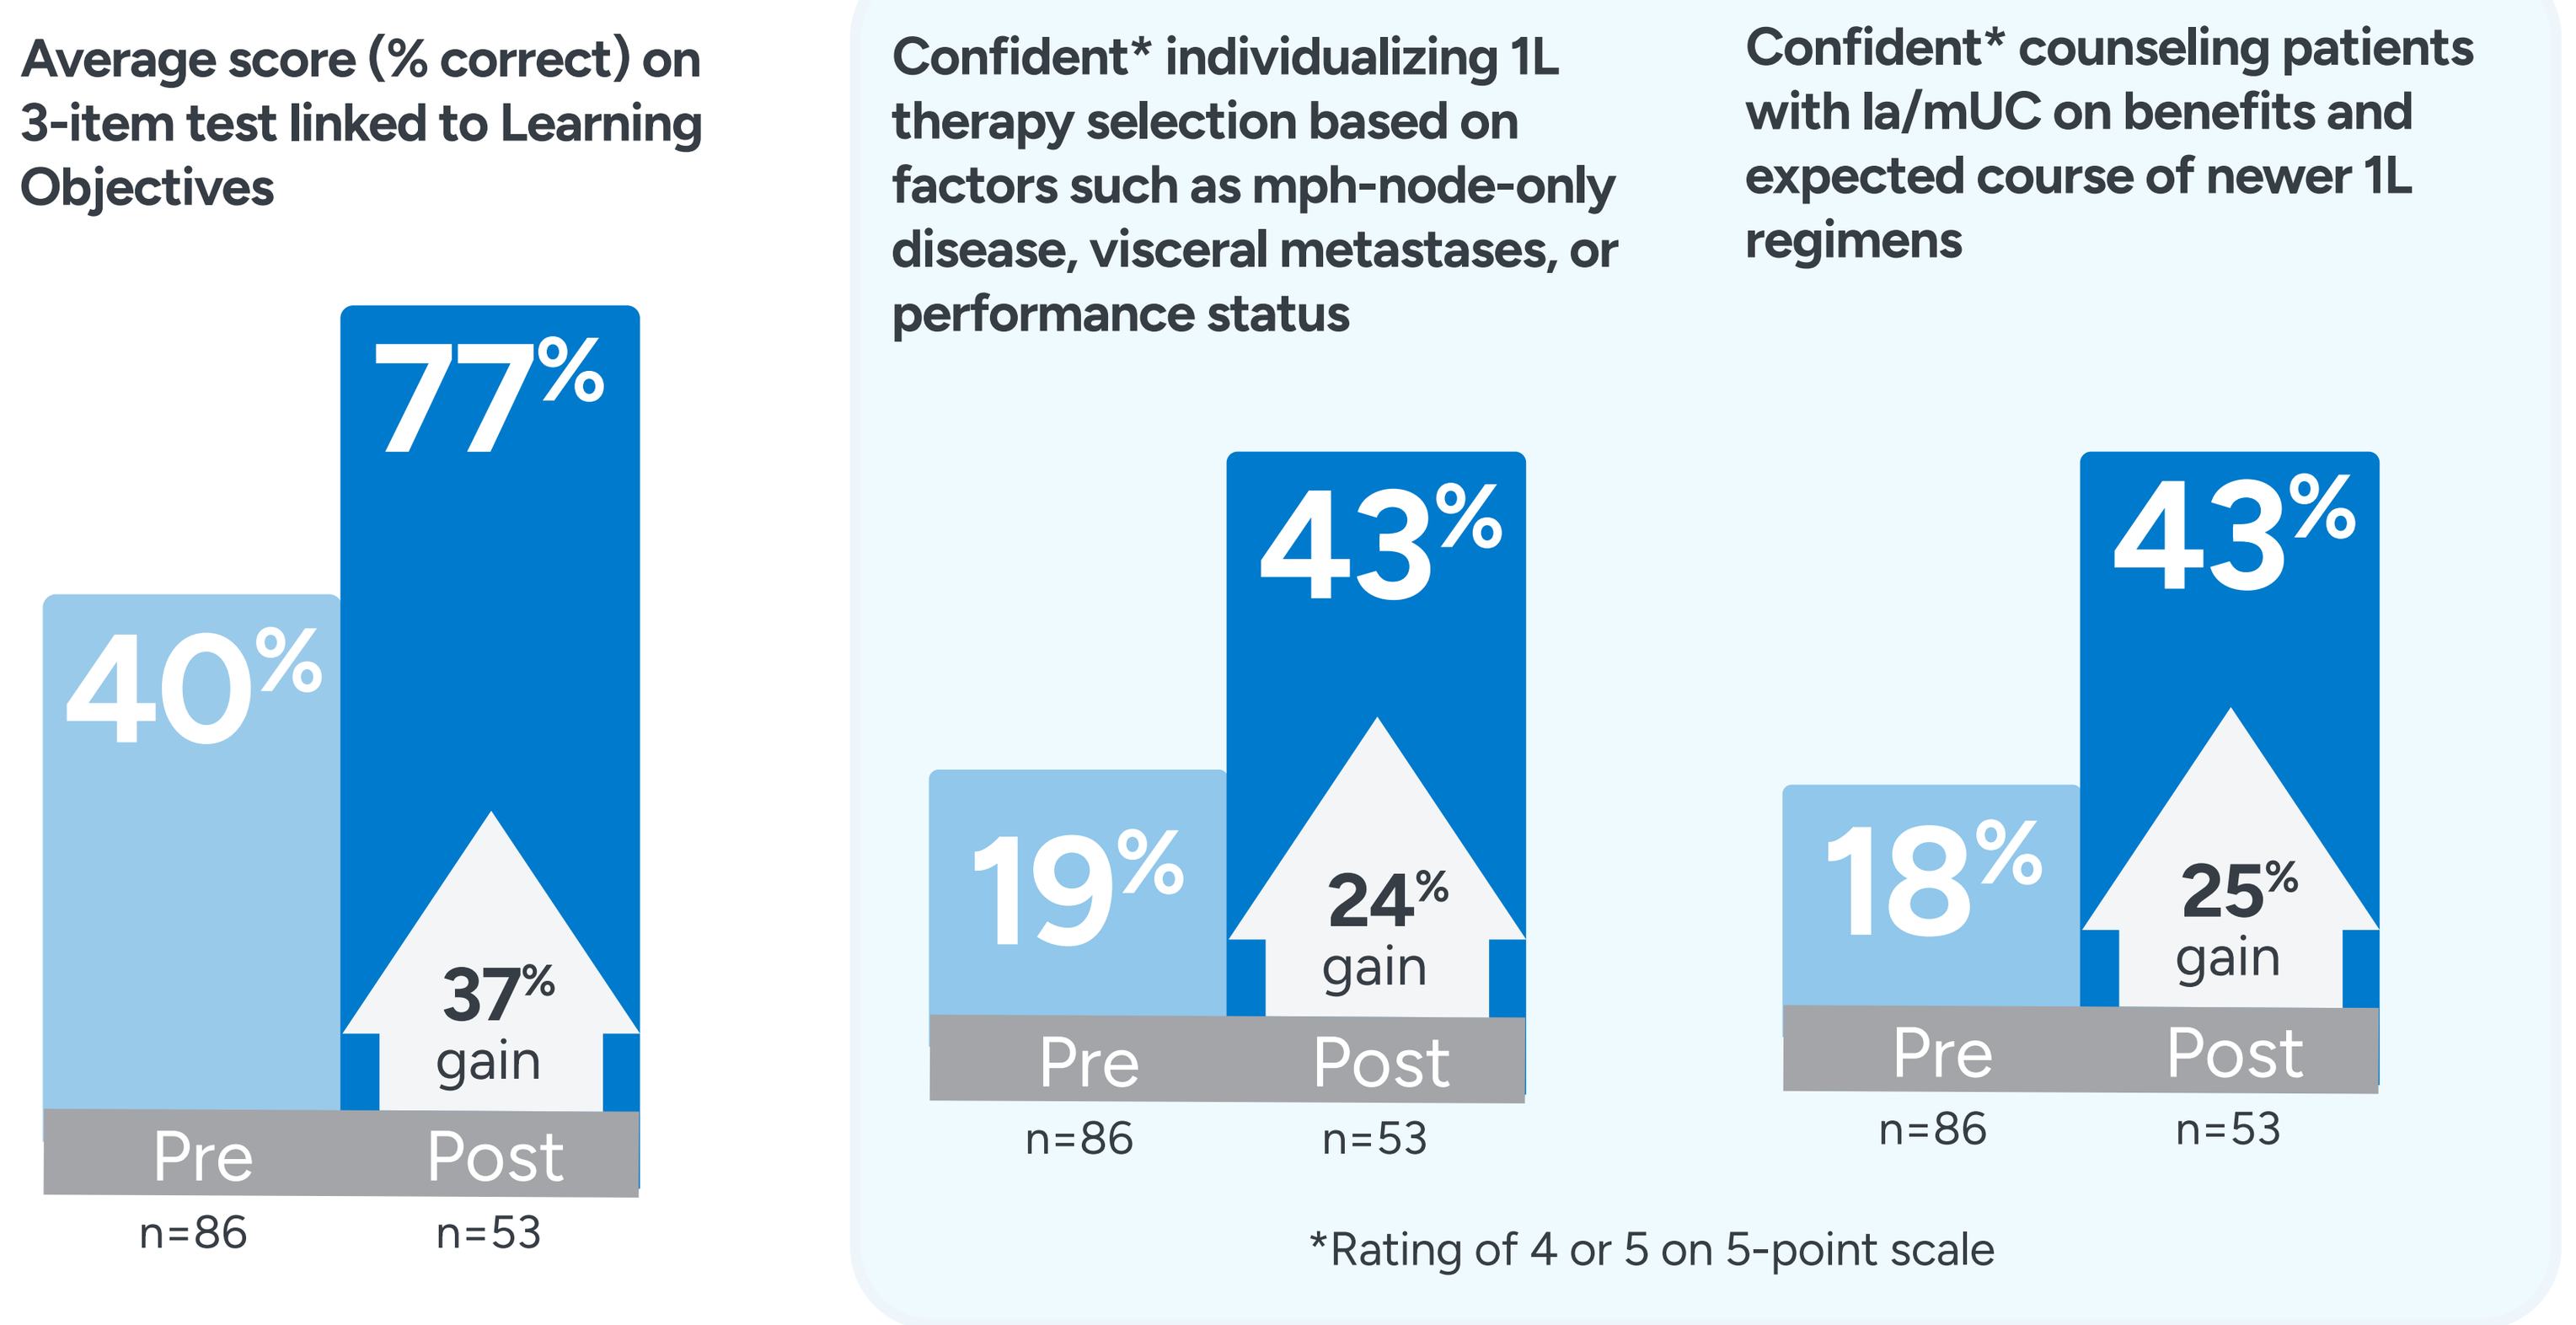
Average score (% correct) on 3-item test linked to Learning Objectives
40%
77%
37% gain
Pre
Post
n=86
n=53
Confident* individualizing 1L therapy selection based on factors such as mph-node-only disease, visceral metastases, or performance status
19%
43%
24% gain
Pre
Post
n=86
n=53
Confident* counseling patients with la/mUC on benefits and expected course of newer 1L regimens
18%
43%
25% gain
Pre
Post
n=86
n=53
*Rating of 4 or 5 on 5-point scale

Following the release of the second CME session (first 3 months of outcomes), the program reached an additional:

7,265 Participants
331 CME and
6,934 micro-learners

78% of CME learners completed the session

71% identified as current treaters seeing an average of 12 patients per month with UC

Intended practice changes for patients with la/mUC (n=160) (Select all that apply)

	Percentage
Strengthen multidisciplinary coordination to streamline ADC+IO initiation	53%
Incorporate enfortumab vedotin + pembrolizumab as a 1L option for eligible patients	46%
Collaborate with financial navigators to address reimbursement and reduce patient financial toxicity	45%
Improve patient counseling on treatment-related toxicities and side-effect management	45%
Implement pharmacy coordination protocols to manage drug access and infusion timing	43%

CONCLUSIONS

- Survey insights and learner-reported challenges were highly concordant, highlighting shared barriers to adoption of guideline-concordant 1L ADC+IO therapy in la/mUC, including eligibility determination, treatment sequencing, and AE management.
- Sequential, survey-informed CME demonstrated broad engagement and improved clinician confidence in first-line la/mUC decision-making.
- Collectively, findings point to a critical need for education that moves beyond awareness to address real-world implementation challenges in clinical practice.
- Both activities continue to accrue learners; ongoing outcomes assessment will further inform educational impact and implications for practice.